CID-078, a first-in-class oral cyclin A/B-RxL inhibitor, elicits anti-tumor activity in breast cancer patient-derived xenograft models

Cristina Molina[1], Andreu Òdena[1,2], Li-Fen Liu[3], Bernard Levin[3], Mariana Paes Dias[1], Marta Guzmán[1], Olga Rodríguez[1], David J. Earp[3], Michael C. Cox[3], Evelyn W. Wang[3], Marie Evangelista[3], Violeta Serra[1].

[1]Vall d'Hebron Institute of Oncology, Barcelona, Spain, [2]University of Barcelona, Barcelona, Spain, [3]Circle Pharma, South San Francisco, CA

Poster Number #P2-05-29

cristinamolina@vhio.net

marie.evangelista@circlepharma.com

1. BACKGROUND

- Cell cycle progression is regulated by cyclins and cyclin-dependent kinases (CDKs), along with various substrates and regulators. E2F activity relies on the interaction of its RxL motif and the hydrophobic patch (HP) on cyclin A. Disrupting this cyclin A/E2F RxL interaction leads to hyperactivation of E2F and **synthetic lethality in E2F-driven tumors**.
- CID-078 is a novel, orally bioavailable, passively cell-permeable, potent, and selective macrocycle that binds to the HP of cyclins A and B **blocking the RxL motif-mediated binding of E2F to cyclin A-CDK2 and Myt1 to cyclin B1-CDK1.** CID-078 induces cell cycle arrest at the G2/M phase, leading to apoptotic tumor cell death.
- Triple negative breast cancer (TNBC) has previously been shown to have higher expression of E2F targets and sensitivity to CID-078 compared to non-TNBC cancer cell lines[1].
- Some ER+/HER2- breast cancer tumors that progress on CDK4/6i may have elevated E2F activity due to alterations in the CDK/RB/E2F pathway[2].
- In lung cancer tumor models, CID-078 treatment induced tumor regression and **increased phosphorylation of separase (ESPL1)**—a mitosis-specific protein and direct substrate of Cyclin B1-Cdk1[1,3].

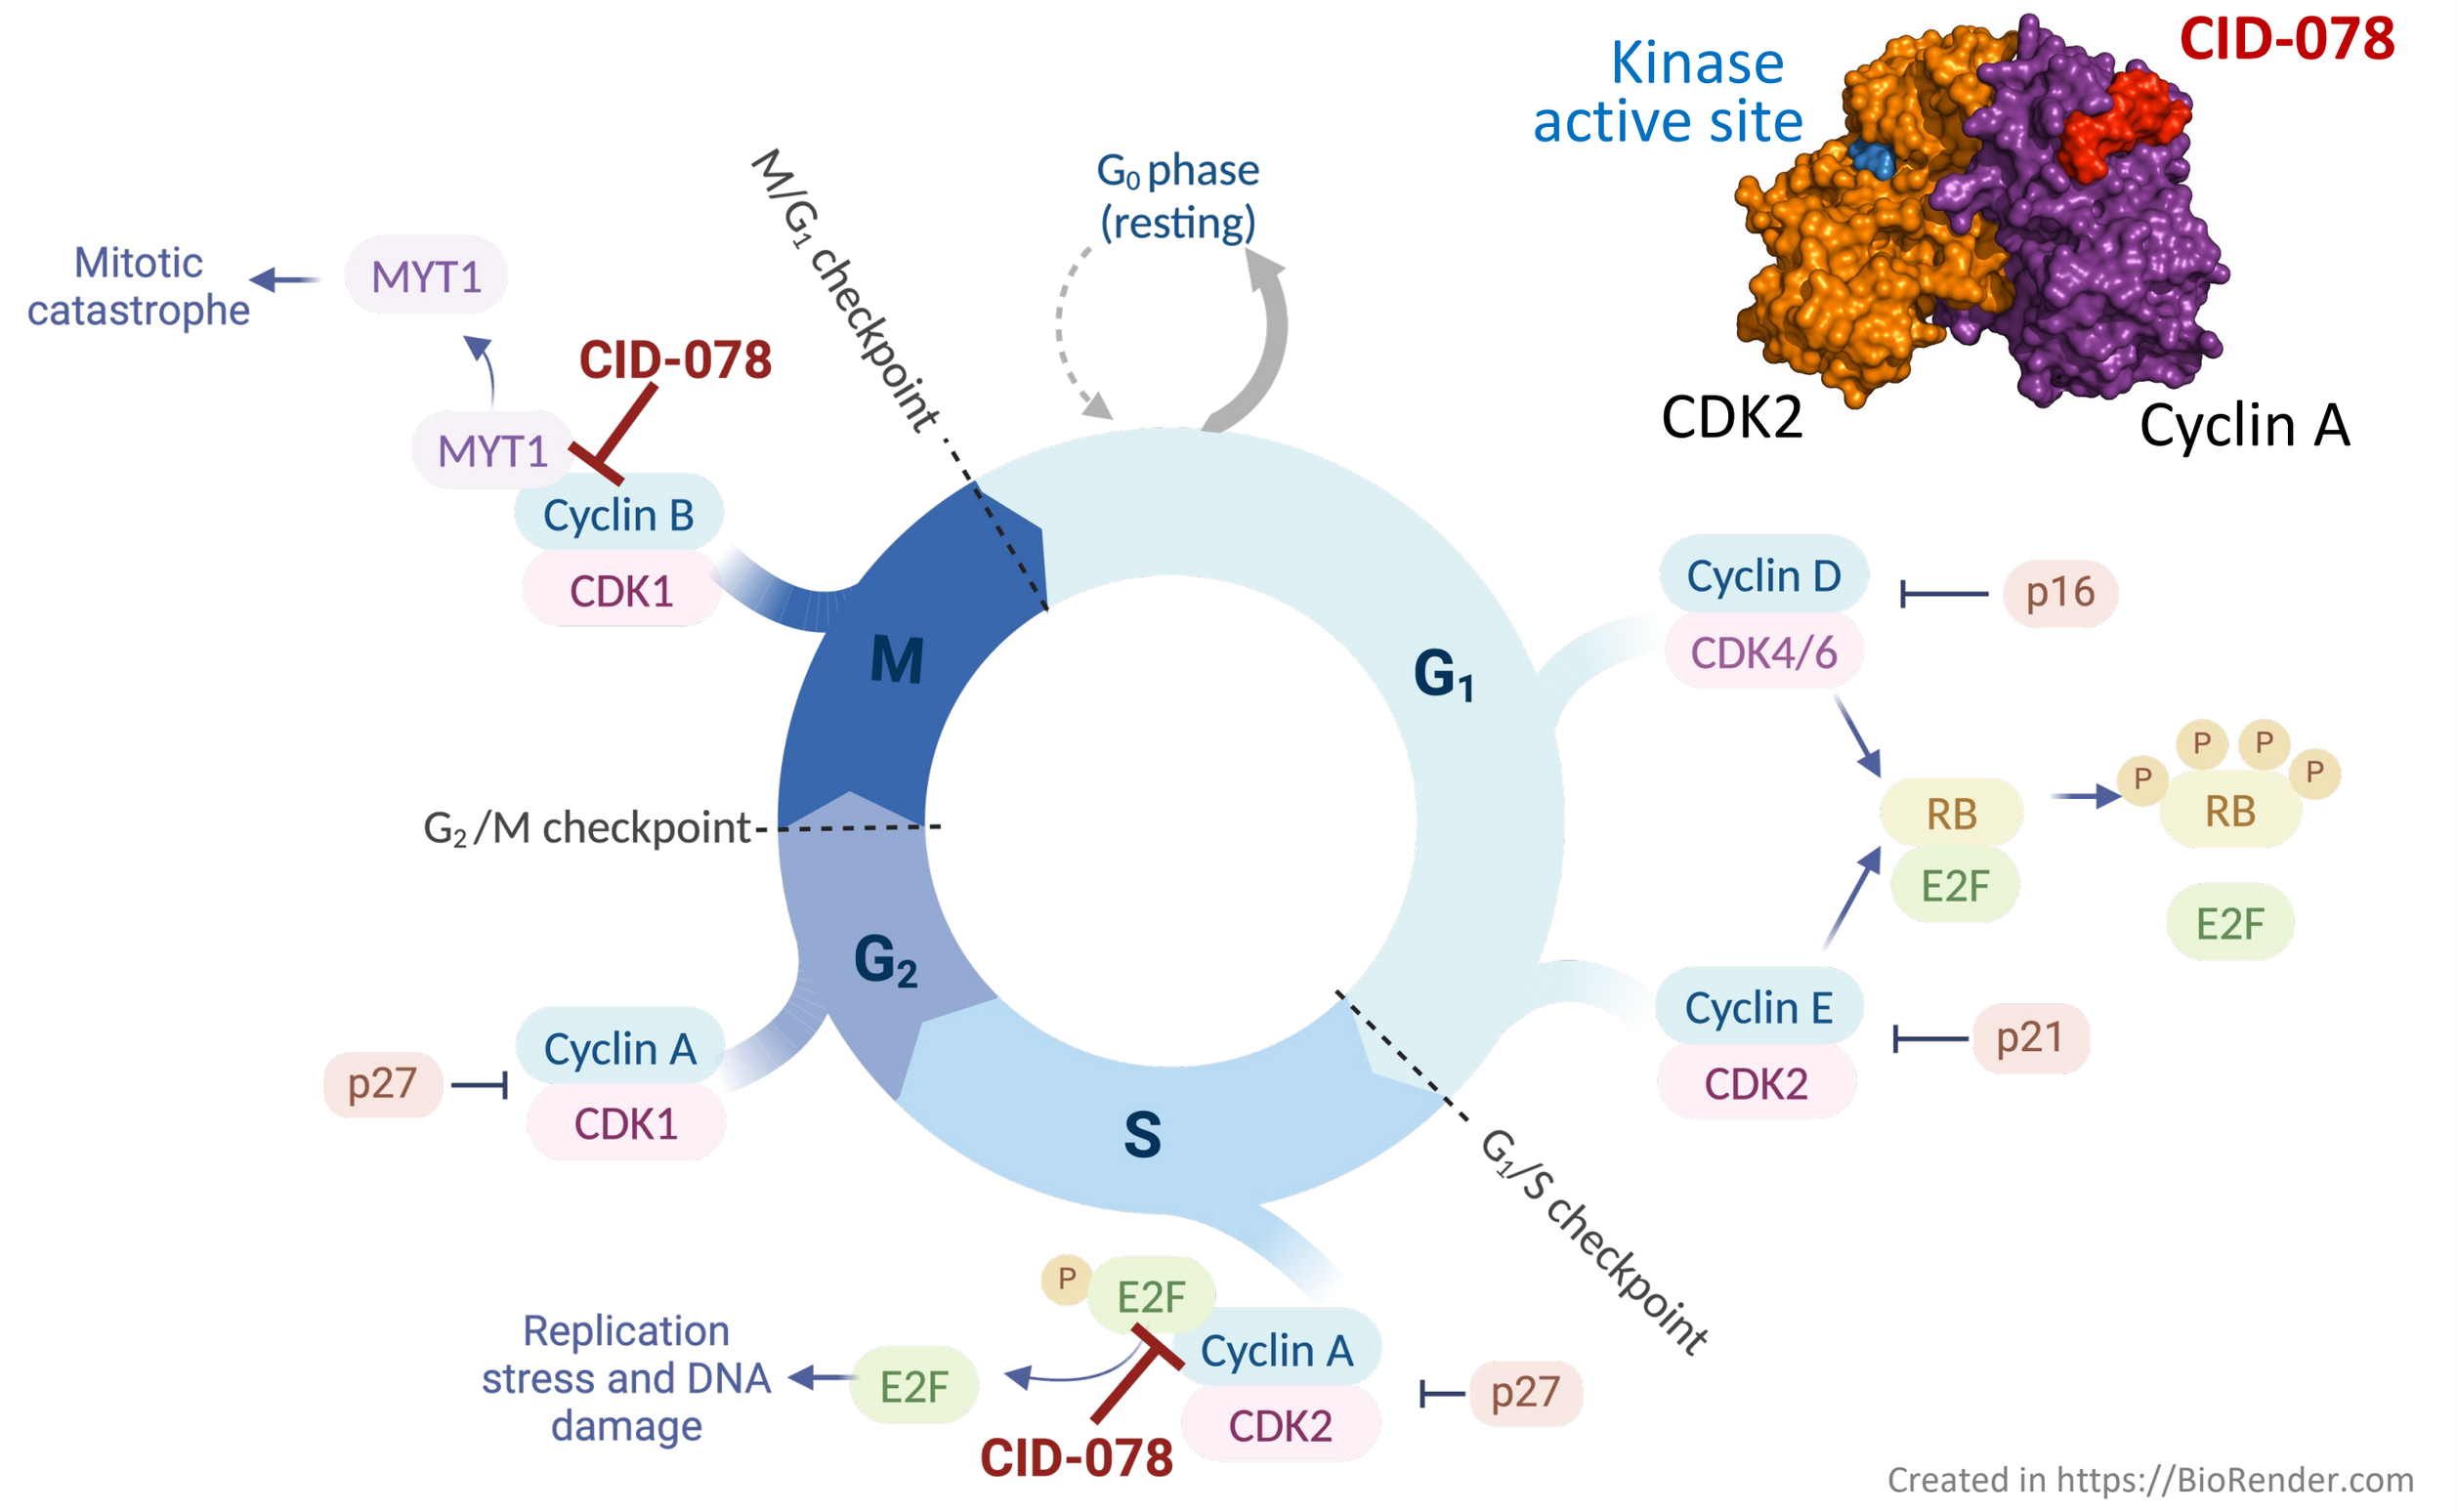
Kinase active site
CID-078
CDK2
Cyclin A
Mitotic catastrophe
MYT1
CID-078
MYT1
Cyclin B
CDK1
M/G1 checkpoint
G0 phase (resting)
M
G1
G2/M checkpoint-
G2
S
Cyclin D
CDK4/6
p16
RB
E2F
P
RB
E2F
Cyclin E
CDK2
p21
p27
Cyclin A
CDK1
G1/S checkpoint
Replication stress and DNA damage
E2F
P
E2F
Cyclin A
CDK2
p27
CID-078
Created in https://BioRender.com

2. AIM

1. Evaluate the anti-tumor efficacy of CID-078 in 8 patient-derived xenografts (PDX) models of breast cancer, including TNBC and ER+/HER2-subtypes.
2. Perform pharmacodynamic evaluation to identify and analyze additional biomarkers associated with response to CID-078.

3. METHODOLOGY

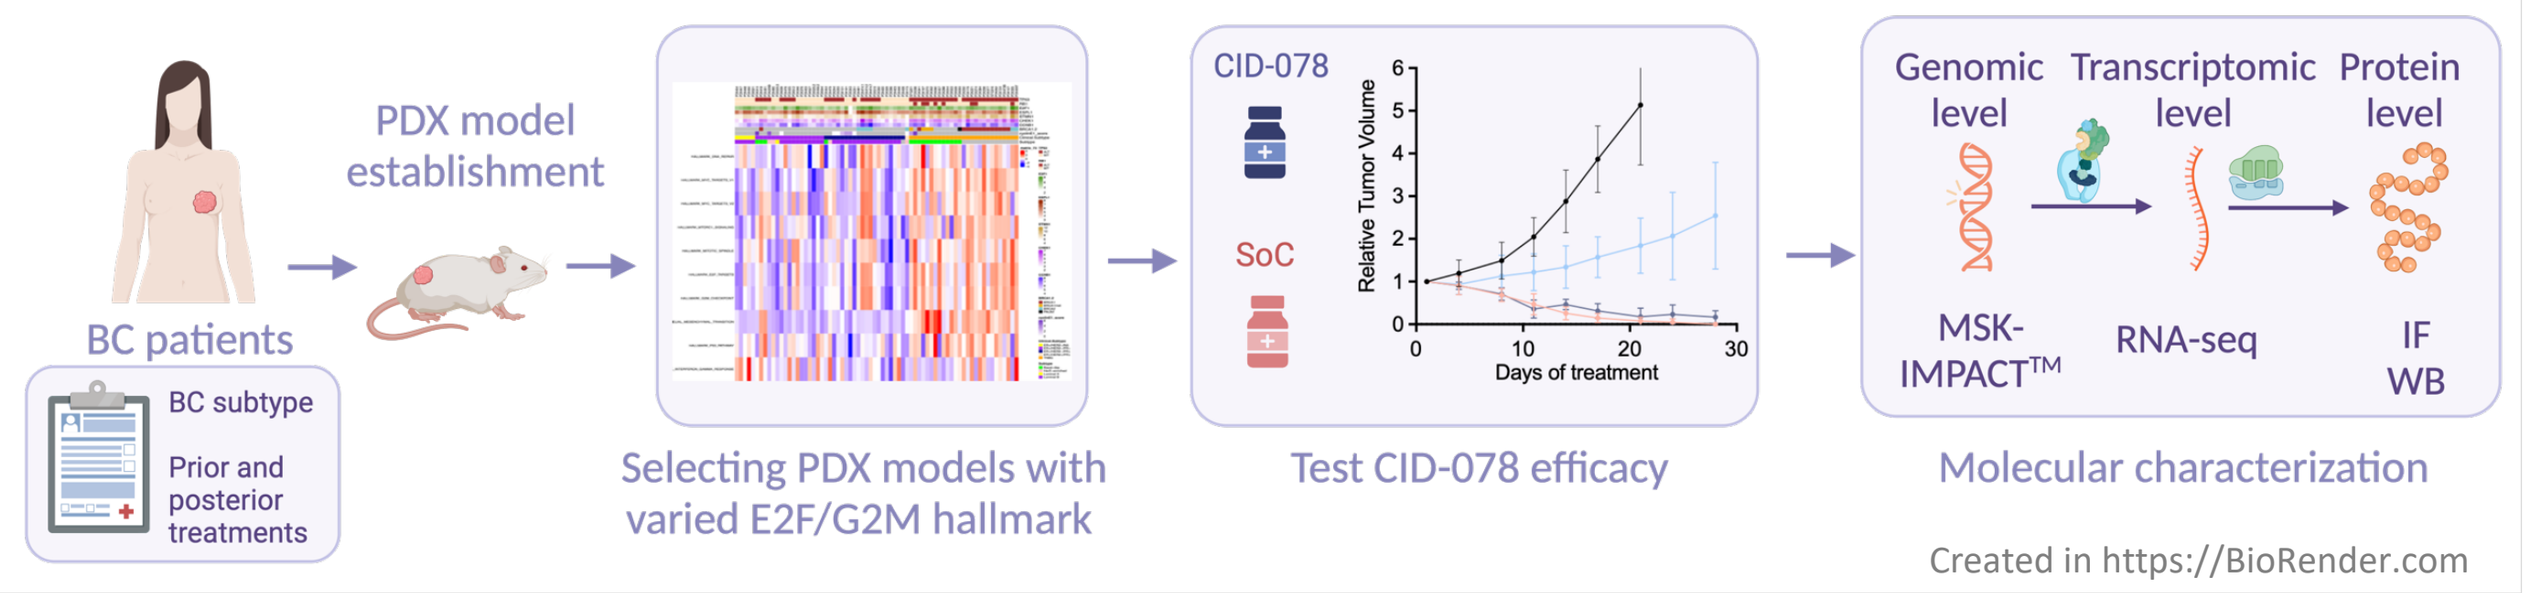
PDX model establishment
BC patients
BC subtype
Prior and posterior treatments
Selecting PDX models with varied E2F/G2M hallmark
CID-078
SoC
Test CID-078 efficacy
Genomic level
Transcriptomic level
Protein level
MSK-IMPACT™
RNA-seq
IF
WB
Molecular characterization
Created in https://BioRender.com

This presentation is the intellectual property of the author/presenter. Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.

4. RESULTS

TNBC PDX models have higher expression of E2F targets; breast cancer models selected for evaluation have varied E2F/G2M hallmark pathways scores

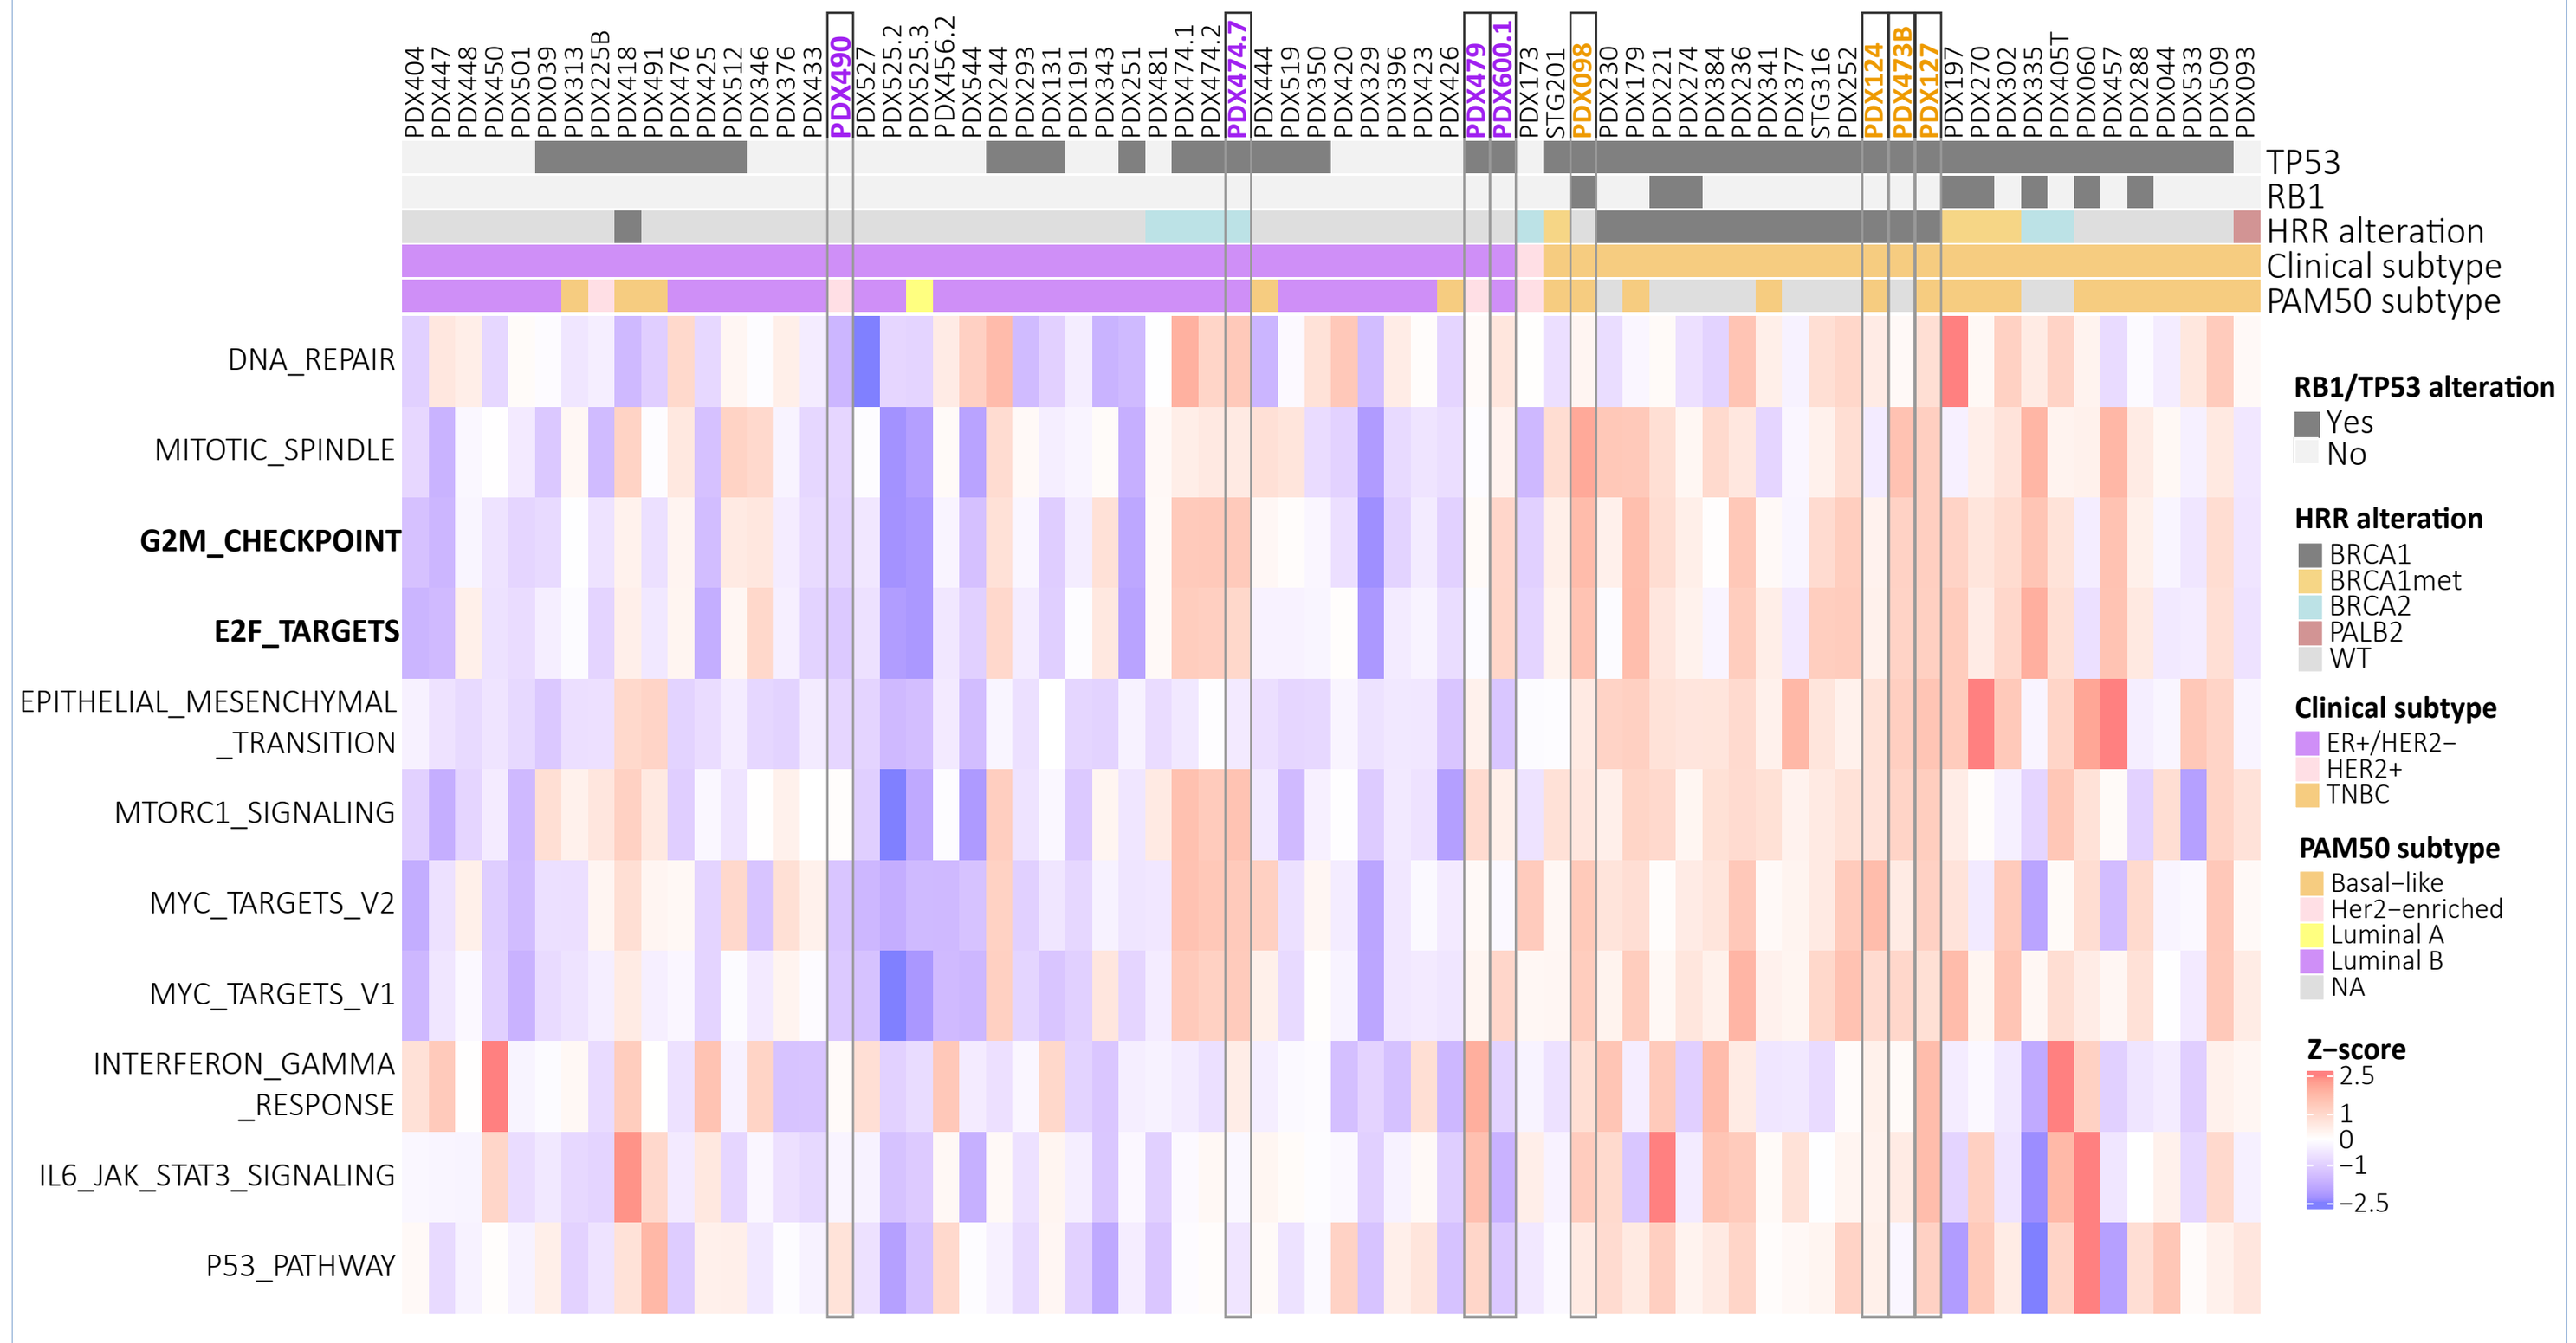
DNA_REPAIR
MITOTIC_SPINDLE
G2M_CHECKPOINT
E2F_TARGETS
EPITHELIAL_MESENCHYMAL_TRANSITION
MTORC1_SIGNALING
MYC_TARGETS_V2
MYC_TARGETS_V1
INTERFERON_GAMMA_RESPONSE
IL6_JAK_STAT3_SIGNALING
P53_PATHWAY
TP53
RB1
HRR alteration
Clinical subtype
PAM50 subtype
RB1/TP53 alteration
Yes
No
HRR alteration
BRCA1
BRCA1met
BRCA2
PALB2
WT
Clinical subtype
ER+/HER2−
HER2+
TNBC
PAM50 subtype
Basal−like
Her2−enriched
Luminal A
Luminal B
NA
Z−score

Figure 1. Heatmap of hallmark pathway scores to select PDX models to test CID-078. A heatmap of hallmark pathway scores was generated for 71 breast cancer patient-derived xenograft (PDX) models, using the GSVA method with the MSigDb Hallmark collection[4,5]. PDX098, PDX124, PDX473B, PDX127, PDX479, PDX600.1, PDX474.7 and PDX490, models which have varied E2F targets and G2M checkpoint hallmark scores, were selected for *in vivo* evaluation.

CID-078 exhibits robust single dose-dependent agent activity in TNBC and post-CDK4/6i ER+/HER2- BC PDX models

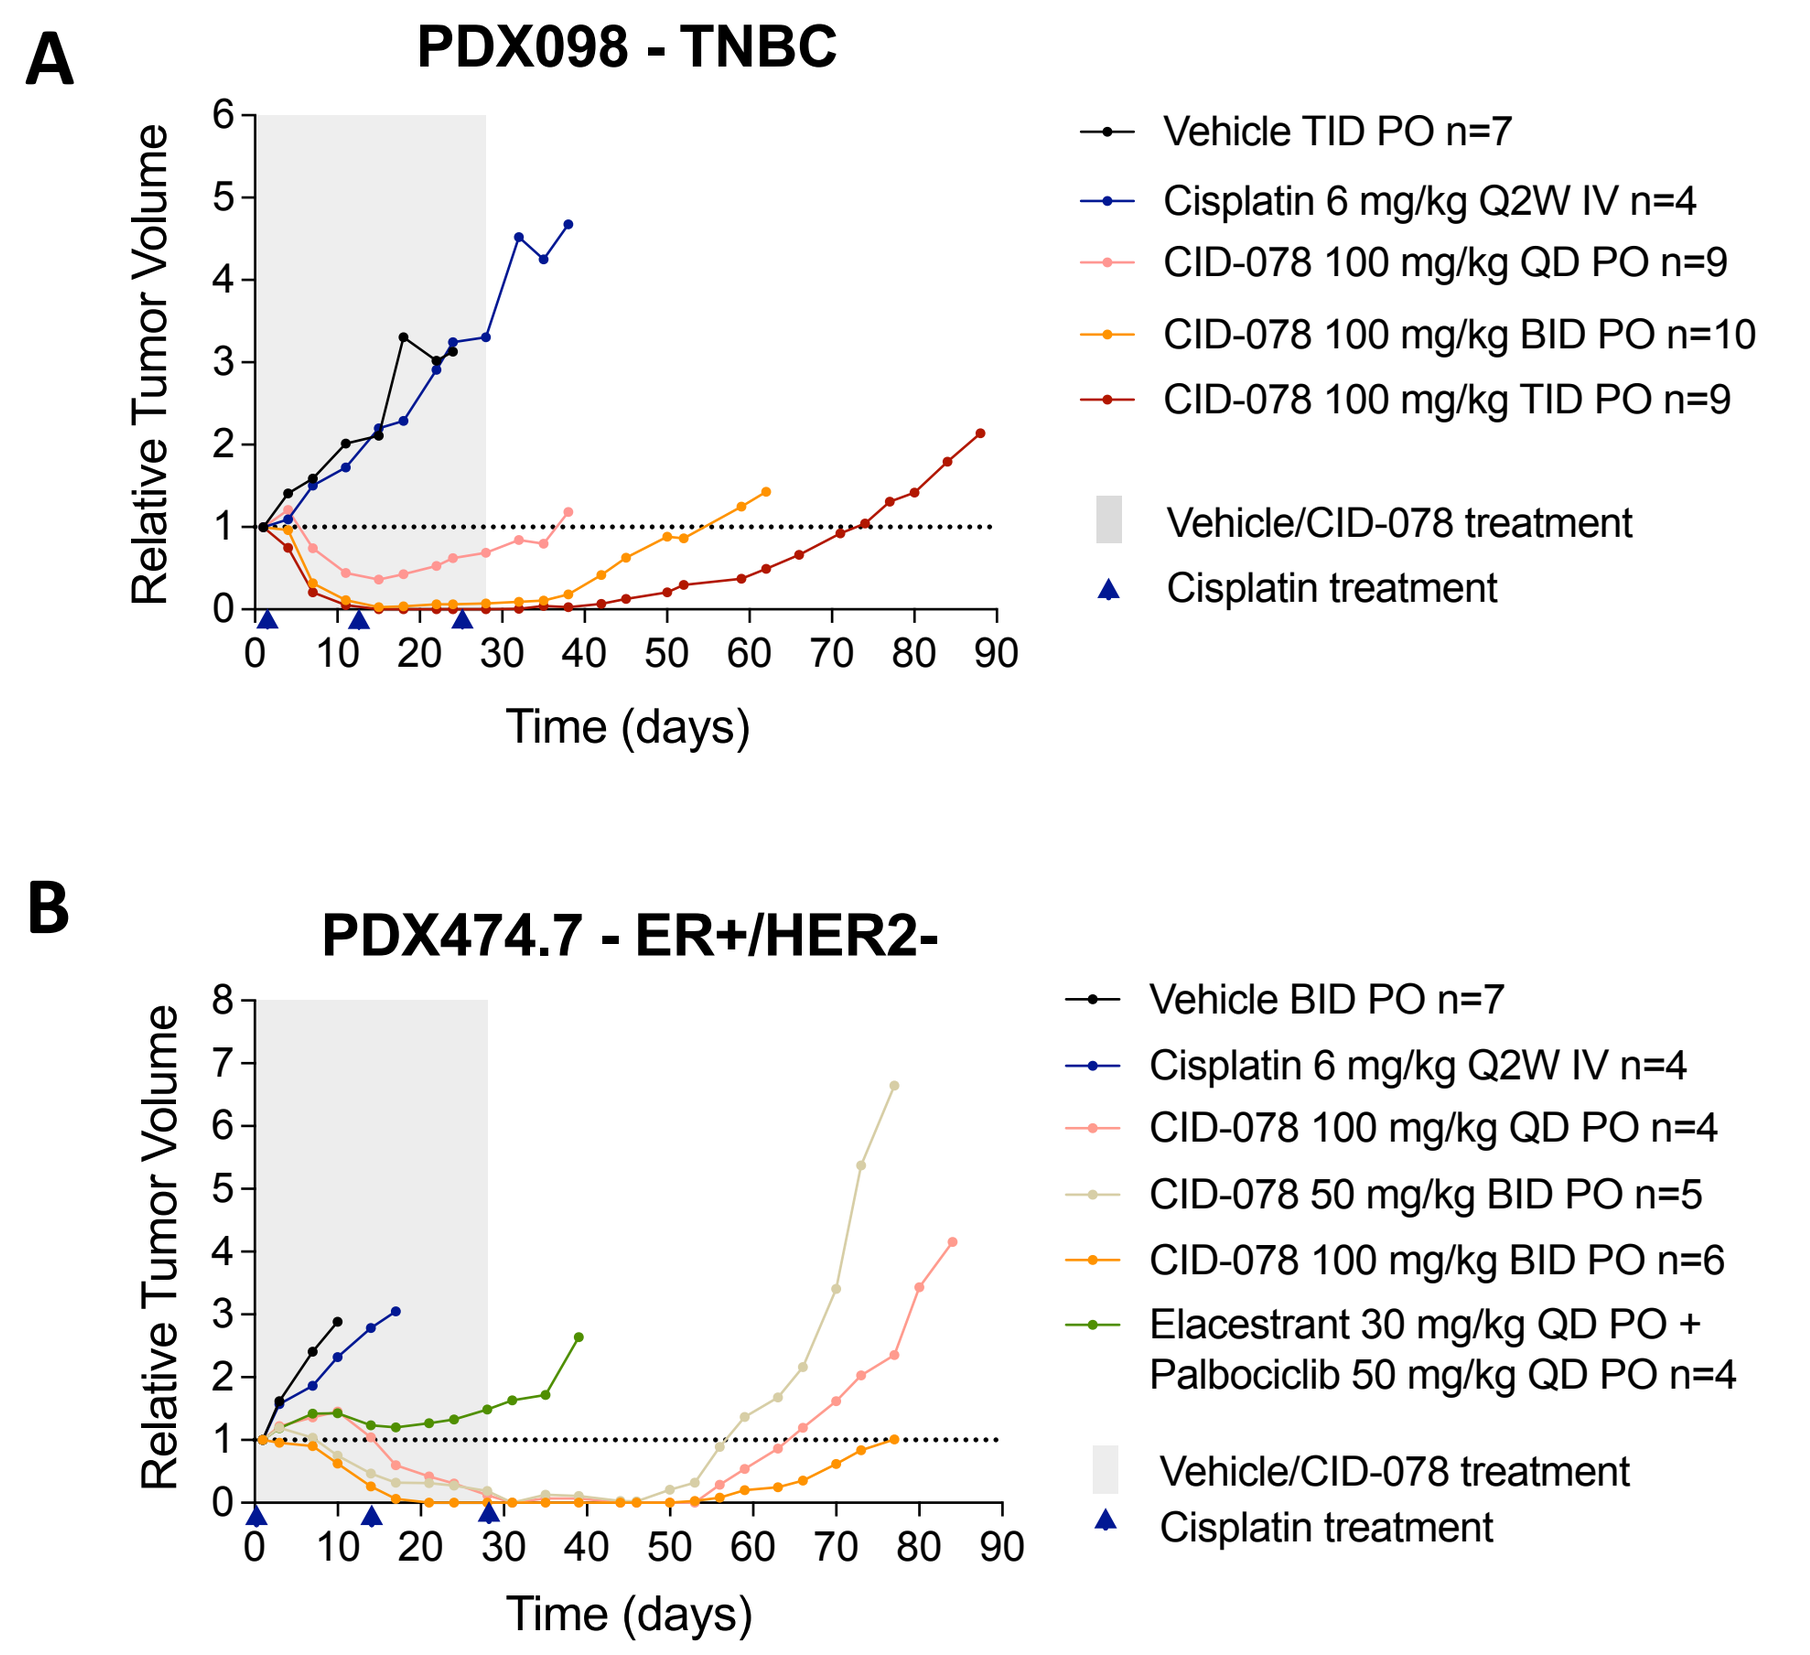
A
PDX098 - TNBC
Vehicle TID PO n=7
Cisplatin 6 mg/kg Q2W IV n=4
CID-078 100 mg/kg QD PO n=9
CID-078 100 mg/kg BID PO n=10
CID-078 100 mg/kg TID PO n=9
Vehicle/CID-078 treatment
Cisplatin treatment
Relative Tumor Volume
Time (days)
B
PDX474.7 - ER+/HER2-
Vehicle BID PO n=7
Cisplatin 6 mg/kg Q2W IV n=4
CID-078 100 mg/kg QD PO n=4
CID-078 50 mg/kg BID PO n=5
CID-078 100 mg/kg BID PO n=6
Elacestrant 30 mg/kg QD PO + Palbociclib 50 mg/kg QD PO n=4
Vehicle/CID-078 treatment
Cisplatin treatment
Relative Tumor Volume
Time (days)

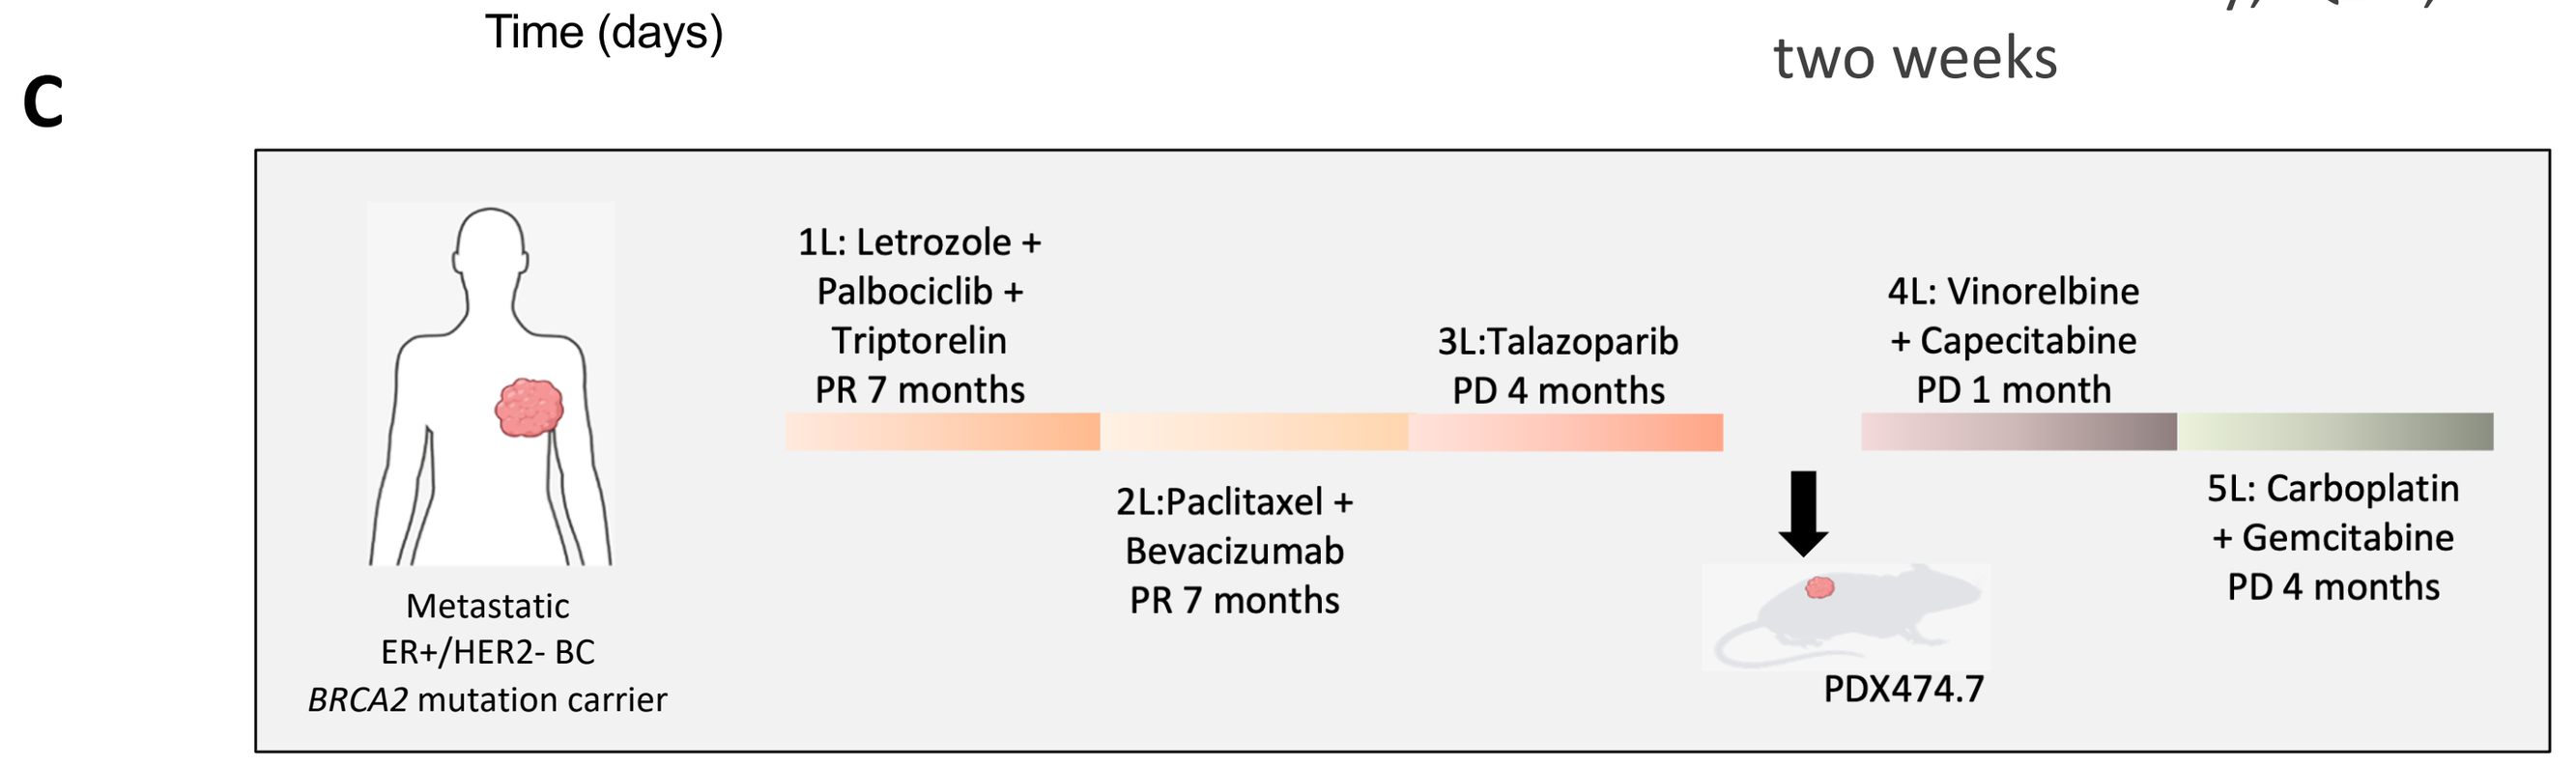
C
Metastatic ER+/HER2- BC BRCA2 mutation carrier
1L: Letrozole + Palbociclib + Triptorelin PR 7 months
2L:Paclitaxel + Bevacizumab PR 7 months
3L:Talazoparib PD 4 months
4L: Vinorelbine + Capecitabine PD 1 month
5L: Carboplatin + Gemcitabine PD 4 months
PDX474.7

Figure 2. NMRI-*Foxn1nu/nu* mice were implanted in the lower flank with tumor fragments of PDX098 (TNBC) **(A)** or PDX474.7 (ER+/HER2- BC) **(B)**. Upon tumor outgrowth (150-250 mm3), mice were treated with CID-078 at indicated doses. Cisplatin was used in both models and endocrine therapy plus a CDK4/6 inhibitor was used for PDX474.7 as a standard-of-care control for comparison. All treatment regimens were tolerated as assessed by body weight measurements (not shown). Patient clinical history for PDX474.7 is shown **(C)**.

PO, orally; IV, intravenously: QD, once daily; BID, twice daily; TID, three times daily; Q2W, once every two weeks

CID-078 anti-tumor activity is associated with higher expression of E2F/G2M hallmark score in breast cancer PDX models

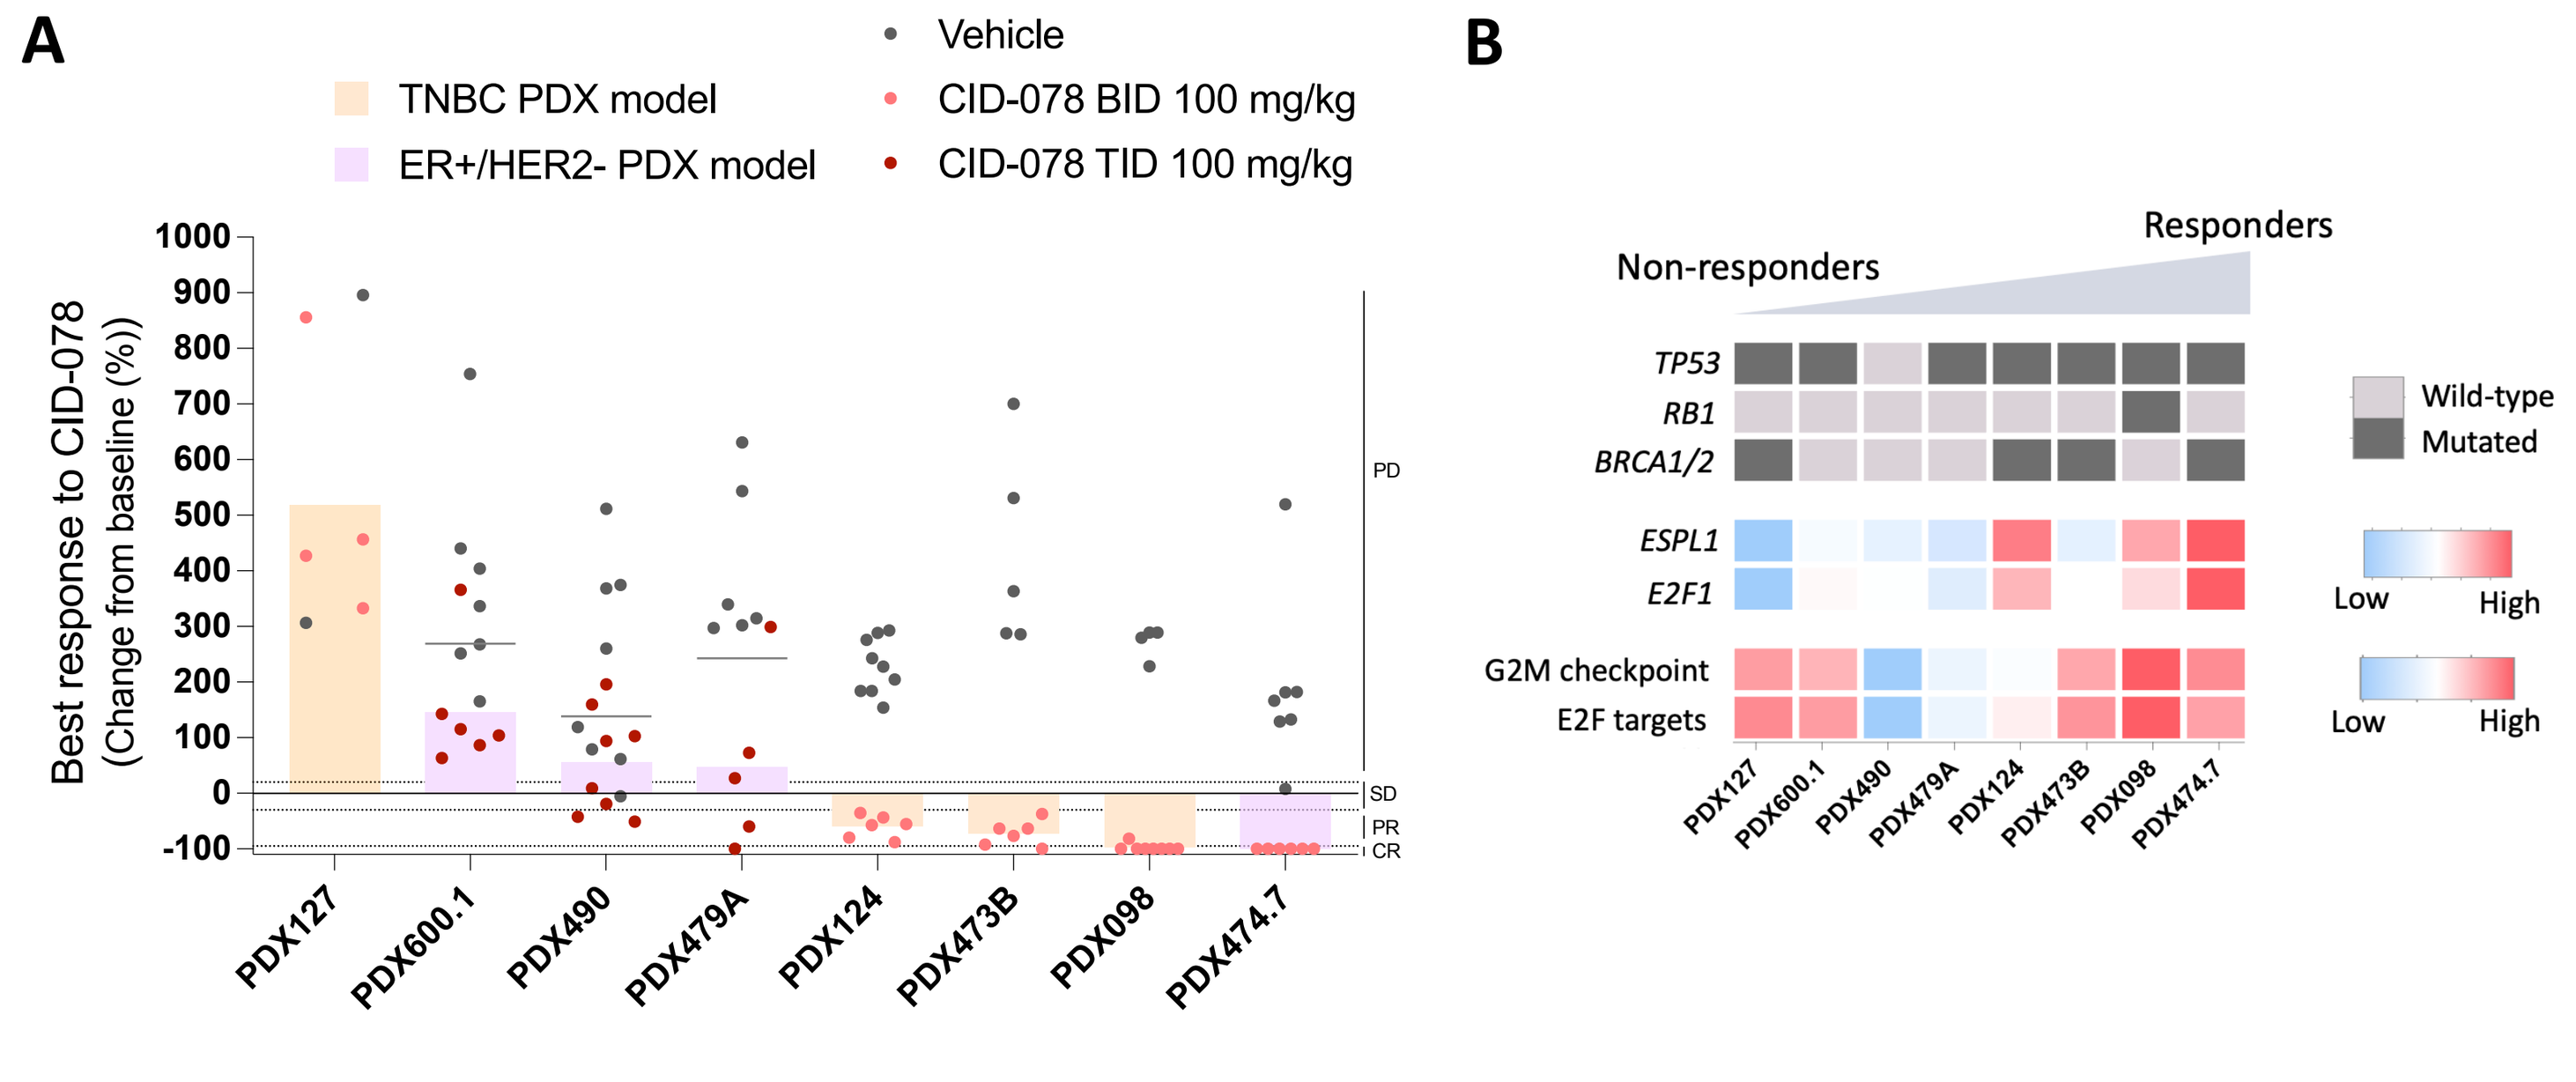
A
TNBC PDX model
ER+/HER2- PDX model
Vehicle
CID-078 BID 100 mg/kg
CID-078 TID 100 mg/kg
Best response to CID-078 (Change from baseline (%))
PD
SD
PR
CR
PDX127
PDX600.1
PDX490
PDX479A
PDX124
PDX473B
PDX098
PDX474.7
B
Non-responders
Responders
TP53
RB1
BRCA1/2
ESPL1
E2F1
G2M checkpoint
E2F targets
Wild-type
Mutated
Low
High

Figure 3. Waterfall plot of *in vivo* efficacy data for CID-078 in breast cancer PDXs models. Best responses from eight BC PDX models **(A)** are shown along with *RB1*, *TP53*, *BRCA1/2* genomic alteration status, *ESPL1* (separase) and *E2F1* gene expression (logCPM), and G2M checkpoint and E2F hallmark pathway scores (Z-score) **(B)**. Of the four ER+/HER2- models tested, PDX490 and PDX479A had not been treated with CDK4/6i, PDX600.1 was refractory to CDK4/6i, and PDX474.7 showed an initial response to CDK4/6i.

High expression of *E2F1*, *E2F2*, and *ESPL1* (separase) correlates with sensitivity to CID-078

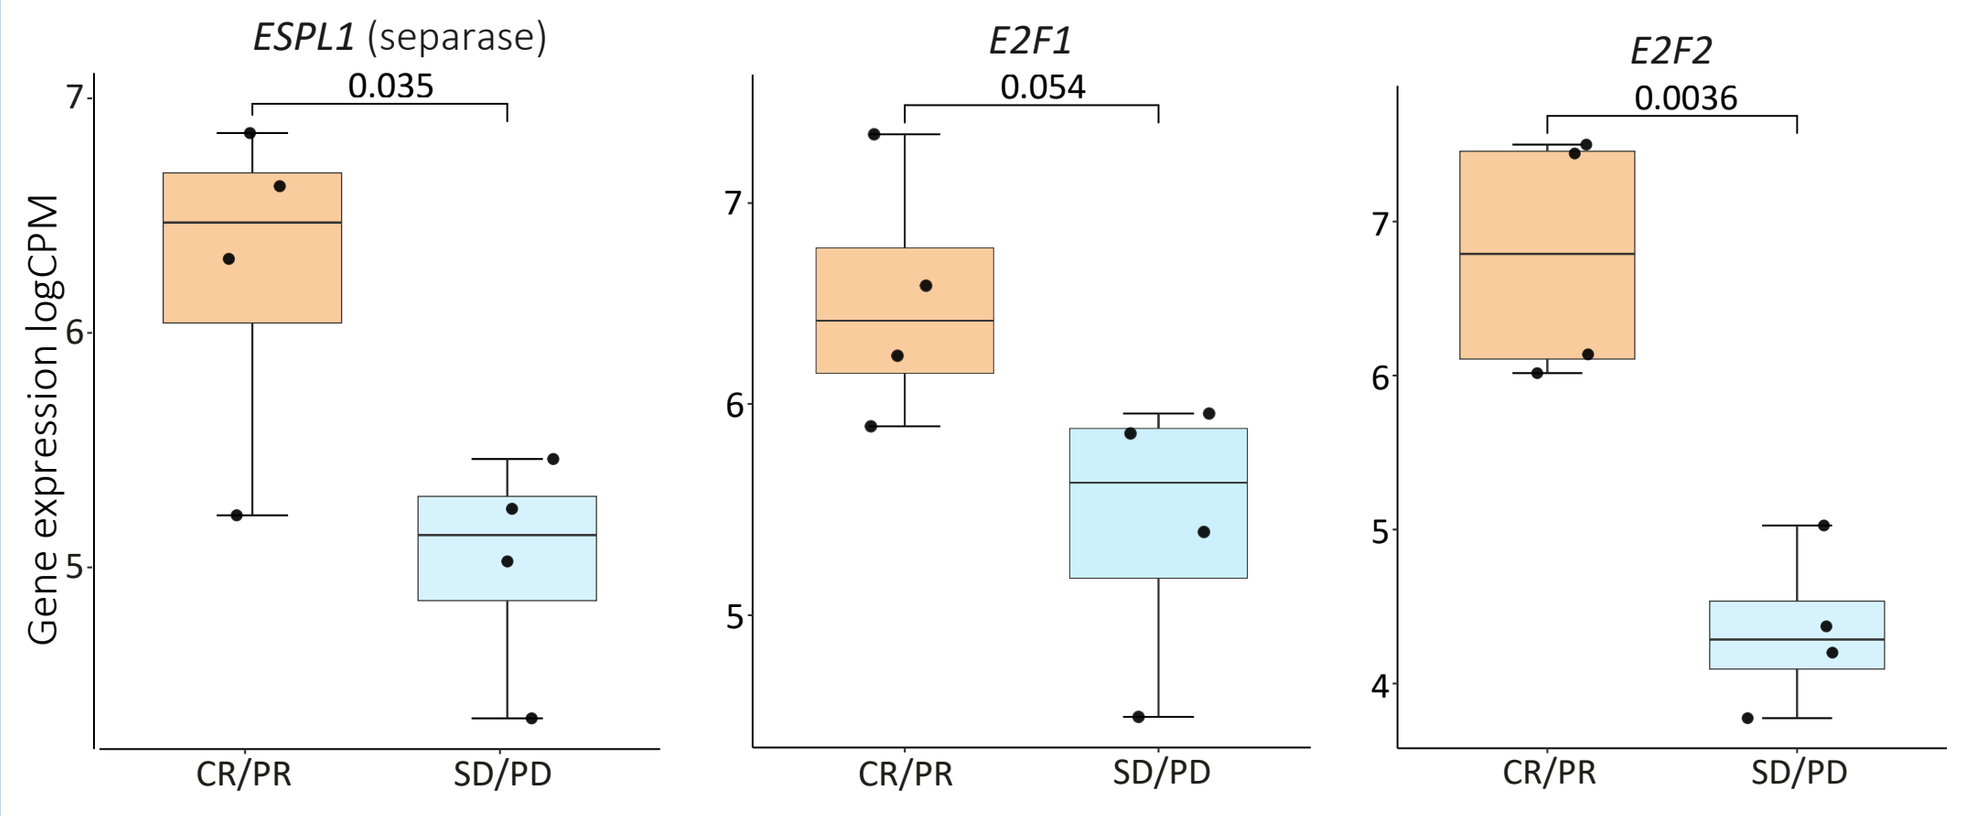
ESPL1 (separase)
0.035
E2F1
0.054
E2F2
0.0036
Gene expression logCPM
CR/PR
SD/PD

Figure 4. Baseline tumor expression levels of *ESPL1*, *E2F1*, and *E2F2*, as measured by RNA-seq, correlate with CID-078 antitumor activity. Statistical significance was determined by Welch's Two Sample t-test.

Treatment with CID-078 induces a robust activation of E2F targets and DNA repair pathways in responders

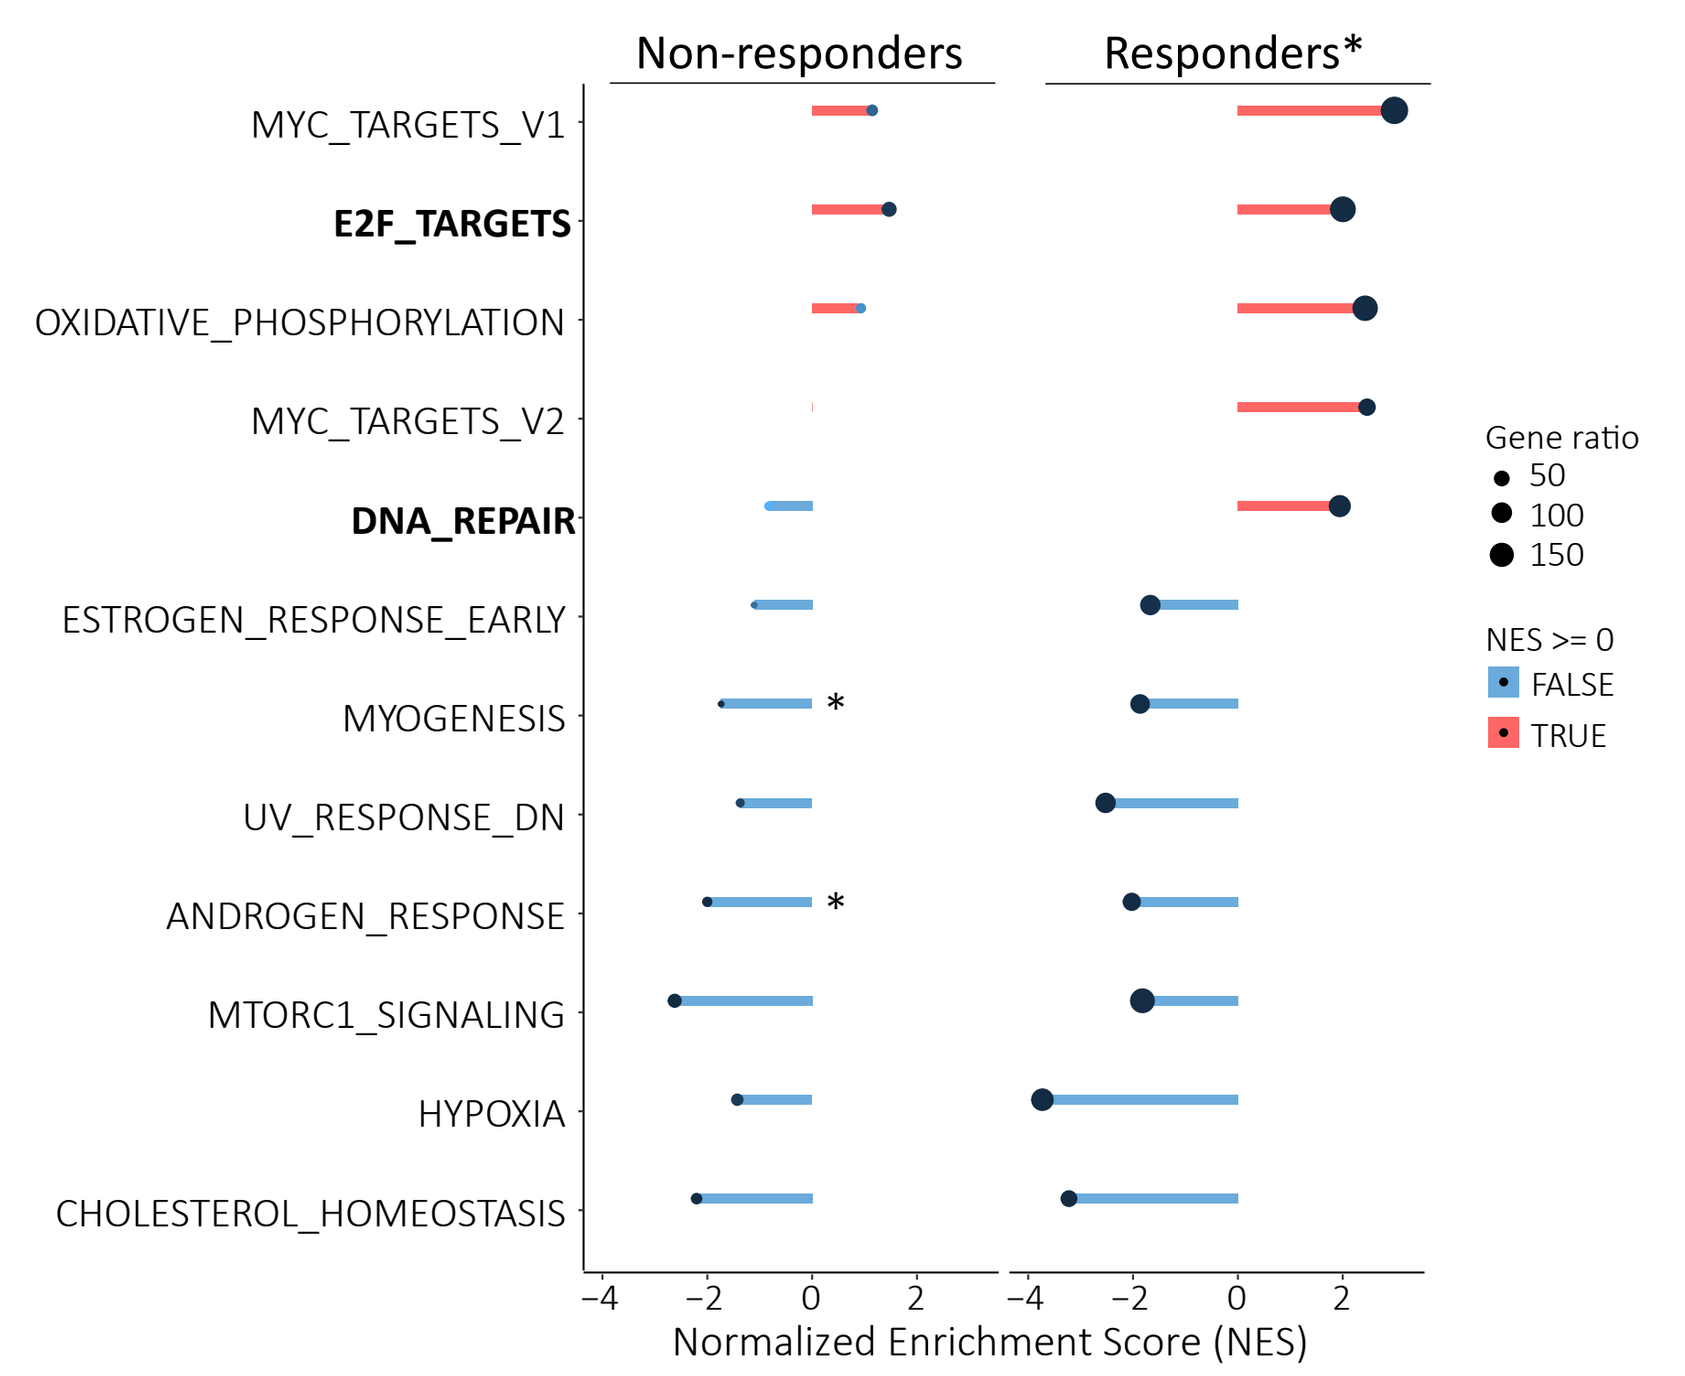
Non-responders
Responders*
MYC_TARGETS_V1
E2F_TARGETS
OXIDATIVE_PHOSPHORYLATION
MYC_TARGETS_V2
DNA_REPAIR
ESTROGEN_RESPONSE_EARLY
MYOGENESIS
UV_RESPONSE_DN
ANDROGEN_RESPONSE
MTORC1_SIGNALING
HYPOXIA
CHOLESTEROL_HOMEOSTASIS
Gene ratio
NES >= 0
FALSE
TRUE
Normalized Enrichment Score (NES)

Figure 5. Gene set enrichment analysis (GSEA) of differentially expressed genes sets between vehicle (n=3-4) and CID-078 treatment (n=3-4) reveals distinct gene set enrichment patterns in non-responders (PDX127 and PDX490, left) and responders (PDX098 and PDX474.7, right). Red and blue lines represent gene sets upregulated and downregulated, respectively, with dot size proportional to the number of enriched genes. All gene sets are significantly enriched in the responder group *p<0.05

Treatment with CID-078 in PDX098 increases mitosis specific biomarker pSeparase (S1126) and DNA damage marker pATM (S1981)

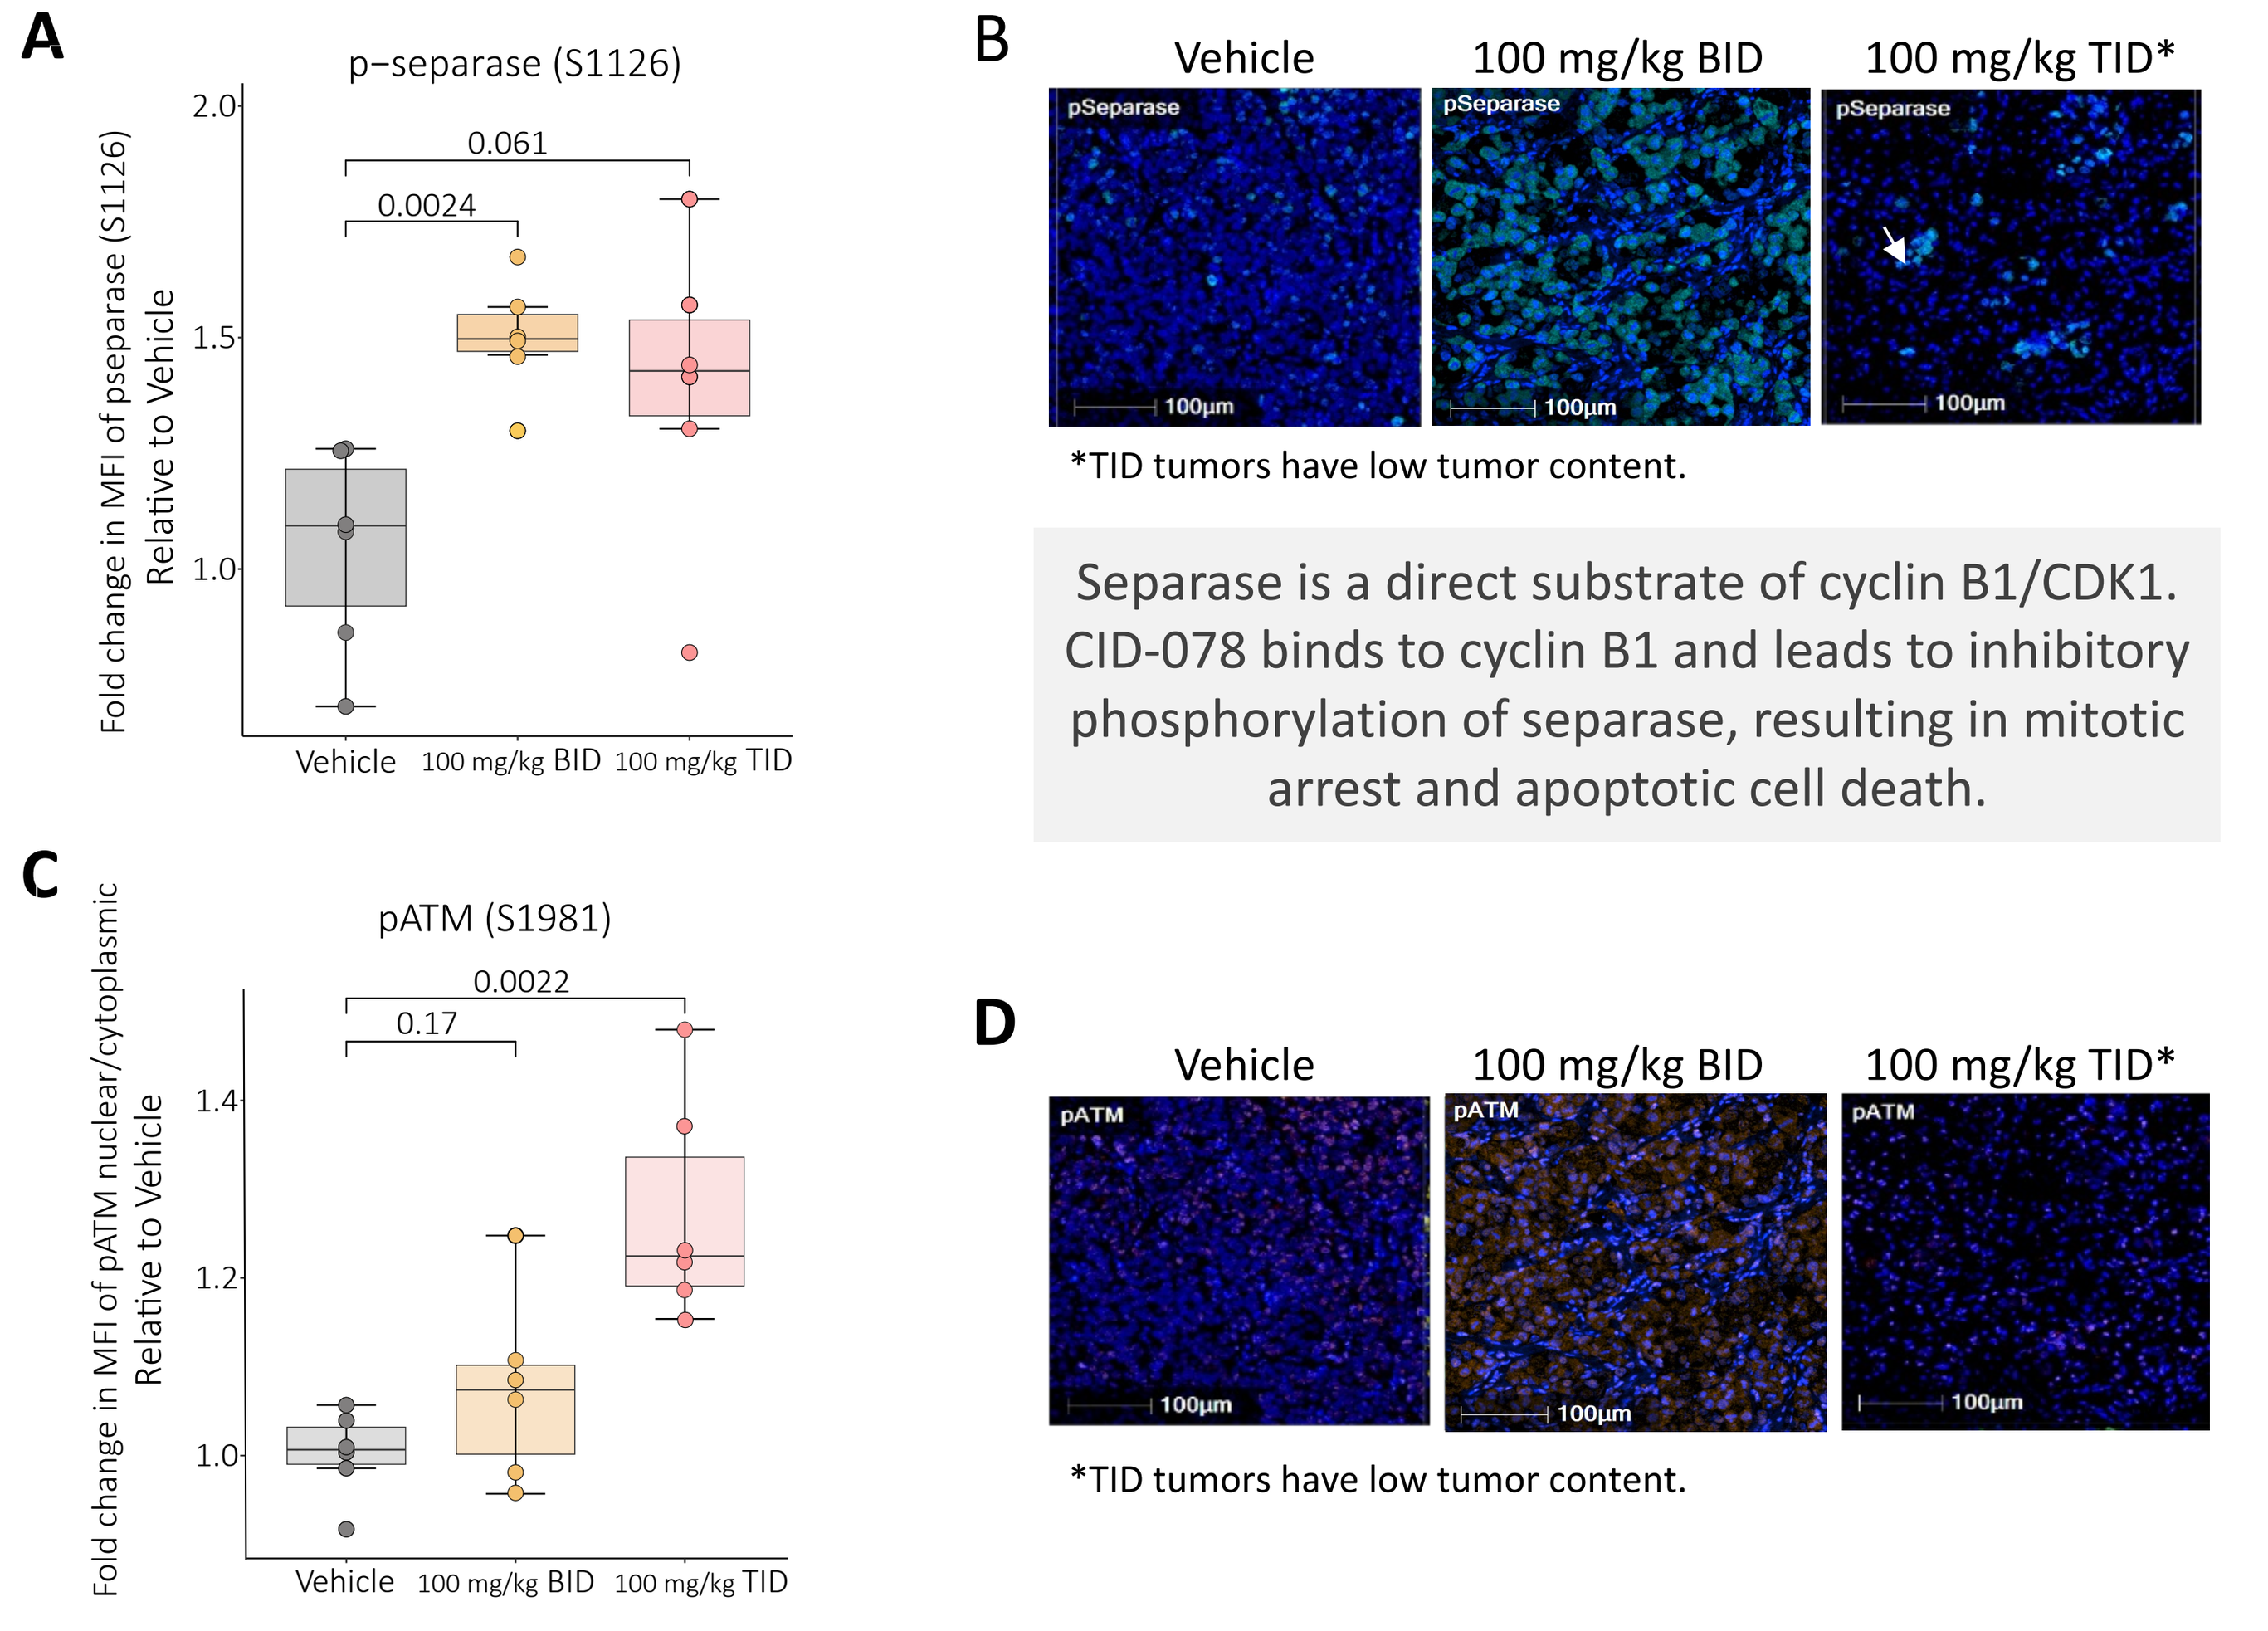
A
p−separase (S1126)
0.061
0.0024
Fold change in MFI of pseparase (S1126) Relative to Vehicle
Vehicle
100 mg/kg BID
100 mg/kg TID
B
Vehicle
100 mg/kg BID
100 mg/kg TID*
pSeparase
100µm
*TID tumors have low tumor content.
Separase is a direct substrate of cyclin B1/CDK1. CID-078 binds to cyclin B1 and leads to inhibitory phosphorylation of separase, resulting in mitotic arrest and apoptotic cell death.
C
pATM (S1981)
0.0022
0.17
Fold change in MFI of pATM nuclear/cytoplasmic Relative to Vehicle
D
pATM
*TID tumors have low tumor content.

Figure 6. Tumor response biomarker modulation observed upon CID-078 treatment in the PDX098 breast cancer model. Representative immunofluorescence (IF) images and quantification of IF results of PDX098 tumors treated with CID-078 show a significant increase in the mean fluorescence intensity (MFI) of pSeparase (S1126) in cyan **(A)** and **(B)** and pATM (S1981) in orange **(C)** and **(D)** compared to treatment with vehicle. MFI was quantified using HaLo and normalized to vehicle-treated controls. Statistical significance was determined by Welch's Two Sample t-test.

5. CONCLUSIONS

- The selective cyclin A/B RxL macrocycle CID-078 exhibits potent antitumor activity in the TNBC and ER+/HER2- breast cancer preclinical models tested, consistent with the proposed mechanism of action and correlating with E2F1 and ESPL1 expression.
- Treatment with CID-078 increases phosphorylation of separase (ESPL1) in sensitive breast cancer PDX models.
- Given these results, CID-078 may be a new treatment option for patients with TNBC or ER+/HER2- breast cancer having progressed through CDK4/6i therapy.
- A multi-center phase 1 clinical trial (NCT06577987) is currently enrolling patients.

6. REFERENCES

[1] Wang E, Liu LF, Levin B, et al. Poster PB-515 presented at the EORTC-NCI-AACR Symposium on Molecular Targets and Cancer Therapeutics, October 23-25, 2024, Barcelona, Spain

[2] Herrera-Abreu MT, Palafox M, Ashgar U, et al. *Cancer Research* 76(8):2301-2313, (2016)

[3] Levin B, Liu LF, Hamkins-Indik F, et al. e-Poster presented at the 2024 World Conference on Lung Cancer, September 7-10, 2024, San Diego, CA

[4] Hänzelmann et al. *BMC Bioinformatics* 14:7, (2016)

[5] Liberzon et al. *Cell Systems* 1, 417-425, (2015)

We would like to acknowledge patients who donated samples for the study and Dr. Sarat Chandarlapaty group from Memorial Sloan Kettering Cancer Center (MSKCC) for the characterization of PDXs model.